A Randomized, Open-Label, Phase II Study of Prophylactic Octreotide (OCT) to Prevent/Reduce the Frequency and Severity of Diarrhea in Patients Receiving Lapatinib (LAP) With Capecitabine (CAP) for the Treatment of Metastatic Breast Cancer (mBC)

319P

Petr Krivorotko,[1] Boguslawa Karaszewska,[2] Stephen Chan,[3] Magdalena Wieczorek-Rutkowska,[4] Tomasz Sarosiek,[5] Marina V Shomova,[6] Elena Ovchinnikova,[7] Juan Pablo Zarate,[8] Chaitali Babanrao Pisal,[9] LaTonya Smith,[8] Alexey Manikhas[10]

[1]Department of Breast Tumors, N. N. Petrov Research Institute of Oncology, Saint Petersburg, Russia; [2]Oncology Outpatient Clinic, The Chemotherapy Clinic and the One Day Chemotherapy Department, Przychodnia Lekarska KOMED, Konin, Poland; [3]Clinical Oncology Department, Nottingham University Hospital NHS Trust (City Hospital Campus), Nottingham, Great Britain; [4]Chemotherapy Department, Copernicus PL Sp.z o.o Wojewódzkie Centrum Onkologii, Gdansk, Poland; [5]Departament of Oncology, Magodent Szpital Elbląska, Warsaw, Poland; [6]Department of Oncology #1, Ryazan Regional Clinical Oncology Center, Ryazan, Russia; [7]City Oncological Dispensary, Nizhny Novgorod Regional Hospital, Nizhny Novgorod, Russia; [8]Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, United States of America; [9]Novartis Healthcare Private Limited, Hyderabad, India; [10]Breast Cancer Department, St. Peterburg City Oncology Dispensary, Saint Petersburg, Russia

Introduction

- LAP in combination with CAP is approved for the treatment of patients with human epidermal growth factor receptor 2 positive (HER2+) breast cancer (BC) whose disease progressed following prior therapy, which included anthracyclines, taxanes and trastuzumab in the metastatic setting.[1,2]
- Diarrhea is the most common adverse event (AE) reported with both LAP and CAP.[3]
- Diarrhea reported with LAP is generally managed with dietary modifications and use of loperamide, which is the recommended first-line treatment option as per the American Society of Clinical Oncology (ASCO) and LAP diarrhea management guidelines.[4,5]
- OCT is approved for the treatment of the severe diarrhea and flushing episodes associated with metastatic carcinoid tumours and for the treatment of diarrhea associated with vasoactive intestinal polypeptide-secreting tumours.[6]
- The present phase II study (NCT02294786) evaluated the clinical benefit of the prophylactic use of OCT to prevent or reduce the frequency and severity of diarrhea associated with LAP+CAP treatment in HER2+ mBC.

Methods

- RELATED was a randomized, parallel-group, multicentre, open-label, phase II study conducted in patients with HER2+ mBC who had progression following prior therapy including anthracyclines, taxanes and trastuzumab in the metastatic setting.
- **Study Treatment:** All patients received LAP (1250 mg once daily [QD]) plus CAP (1000 mg/m^2 twice daily [BID]) until disease progression. Patients were randomized in a 1:1 ratio to receive either OCT (Sandostatin® long-acting release [LAR] formulation) or no OCT treatment (**Figure 1**)
 - Patients received OCT 40 mg 7 days prior to the start of LAP+CAP treatment and again 28 days later
 - For patients who received no OCT, LAP+CAP treatment was started immediately after enrolment

Figure 1. Study Design

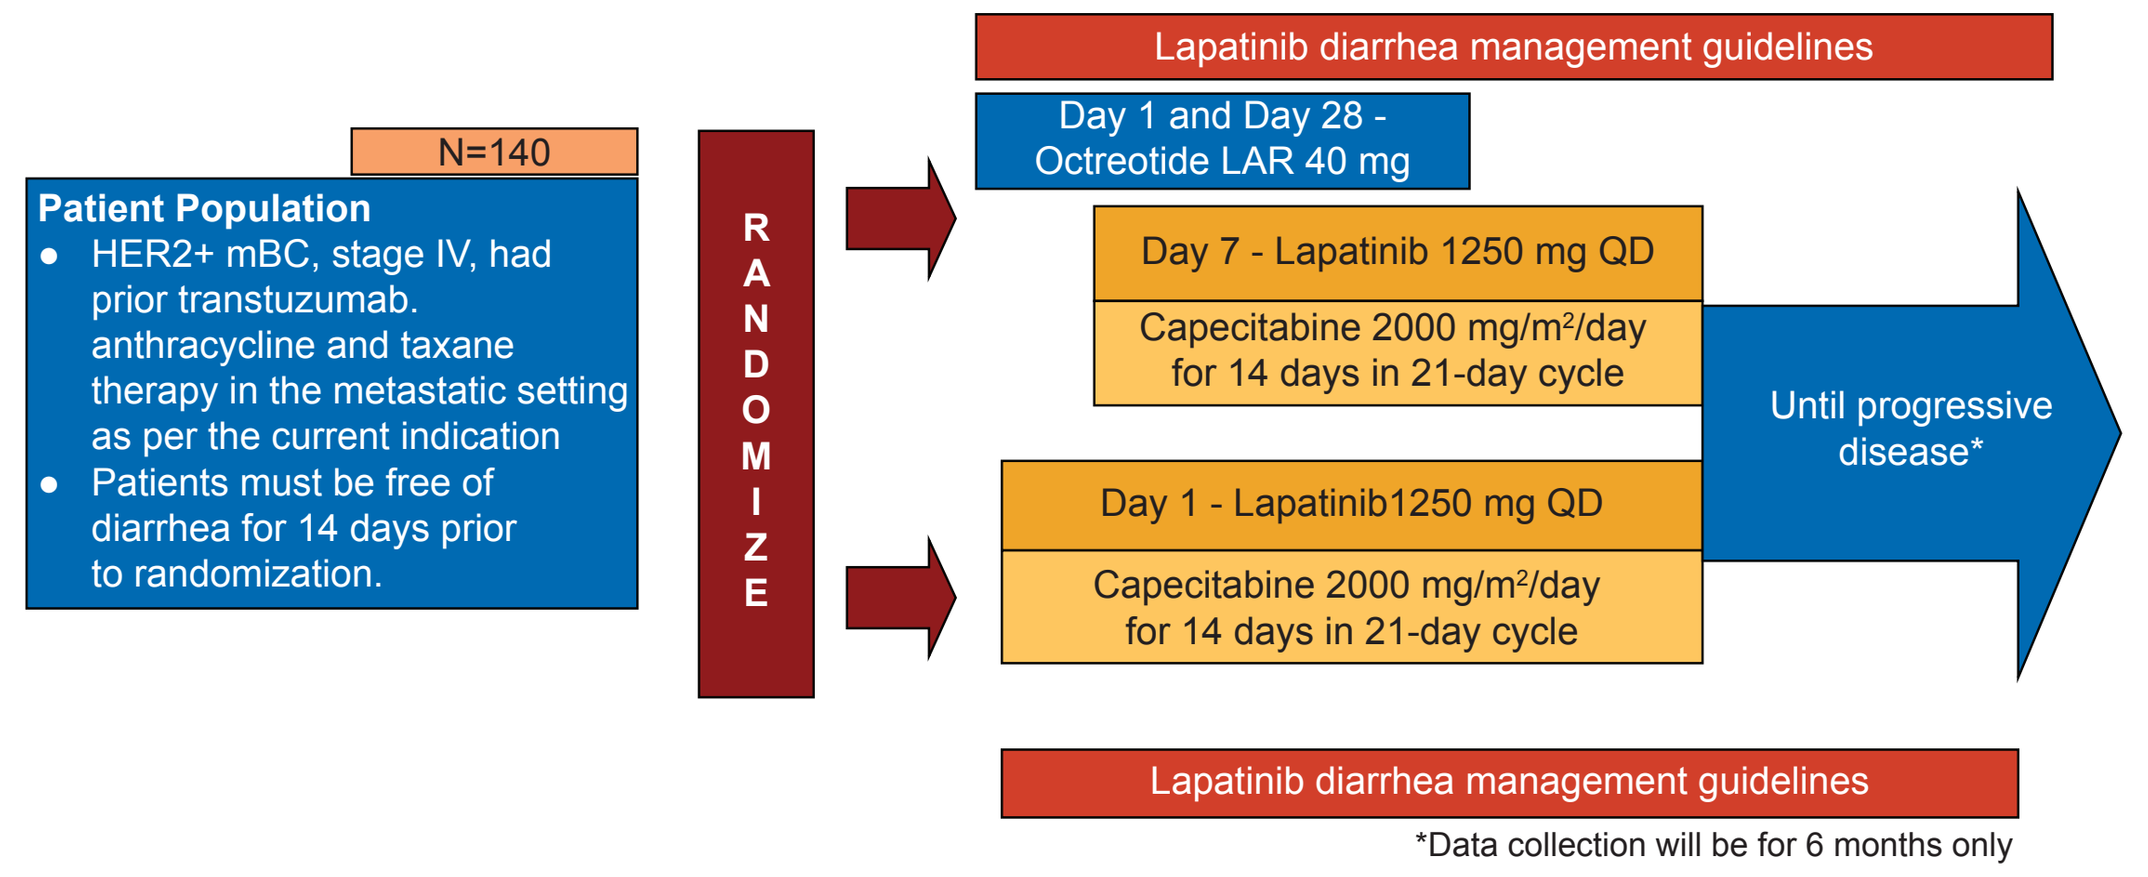

Lapatinib diarrhea management guidelines
N=140
Patient Population
HER2+ mBC, stage IV, had prior transtuzumab. anthracycline and taxane therapy in the metastatic setting as per the current indication
Patients must be free of diarrhea for 14 days prior to randomization.
RANDOMIZE
Day 1 and Day 28 - Octreotide LAR 40 mg
Day 7 - Lapatinib 1250 mg QD
Capecitabine 2000 mg/m²/day for 14 days in 21-day cycle
Day 1 - Lapatinib1250 mg QD
Capecitabine 2000 mg/m²/day for 14 days in 21-day cycle
Until progressive disease*
Lapatinib diarrhea management guidelines
*Data collection will be for 6 months only

HER2+, human epidermal growth factor receptor 2 positive; LAR, long-acting release; mBC, metastatic breast cancer; QD, once daily

Key Eligibility Criteria

- The key inclusion and exclusion criteria for all patients are described in **Table 1**.

Table 1. Key eligibility Criteria

Inclusion Criteria	Exclusion Criteria
• Adult women aged ≥18 years with histologically or cytologically confirmed HER2+ mBC whose disease progressed following prior therapy which included anthracyclines, taxanes and trastuzumab in the metastatic setting	• Concurrent treatment with an investigational agent or participation in any another clinical trial was not allowed
• ECOG performance status 0–2	• OCT treatment within 3 months prior to randomization and prior treatment with LAP
• Completion of FACIT-D and diarrhea diary before receiving the first dose of OCT if randomized to receive OCT	• Concurrent chemotherapy, radiotherapy, immunotherapy, biologic therapy, or hormonal therapy for cancer was not permitted
• Prior treatment with chemotherapy or endocrine therapy was allowed	• Patients with diarrhea (any grade) within 14 days prior to treatment with OCT for patients randomized to receive OCT or within 14 days prior to the first dose of LAP+CAP for those randomized to receive no OCT
• Prior radiation therapy at least 2 weeks before initiating treatment with OCT is permitted	• Patients with malabsorption syndrome, inflammatory bowel disease (ulcerative colitis, Crohn's disease), irritable bowel syndrome, disease significantly affecting the gastrointestinal function or resection of the stomach or small bowel

ECOG, Eastern Cooperative Oncology Group; FACIT-D, Functional Assessment of Chronic Illness Therapy-Diarrhea; HER2+, human epidermal growth factor receptor 2 positive; LAP, lapatinib; mBC, metastatic breast cancer; OCT, octreotide

Study Objectives

- **Primary objective**: To determine the efficacy of prophylactic OCT in reducing the proportion of patients experiencing diarrhea with a severity of the National Cancer Institute Common Terminology Criteria for Adverse Events (NCI CTCAE [version 4.03]) grade ≥2.
- **Secondary objectives**: To determine the efficacy of prophylactic OCT for the following endpoints:
 - In reducing the proportion of patients experiencing diarrhea with a severity of any grade and grade ≥3
 - In increasing the time to onset of the first episode of diarrhea of any grade of severity
 - Compliance with LAP and CAP treatment
 - Efficacy of treatment with LAP and CAP in terms of overall response rate (ORR) and clinical benefit response (CBR)
 - Safety and tolerability of the combination

Statistical Analysis

- Assuming an incidence of 32% of grade ≥2 severity diarrhea for patients who receive LAP+CAP with no prophylactic OCT, using a 1-sided test with an alpha level of 0.025, 140 patients were needed to detect a reduction of 20% in the incidence of grade ≥2 diarrhea, with >80% power.
- An interim analysis (IA) was planned when at least 60 patients were randomized to treatment and completed the 9-week assessment or discontinued from the study. Based on the IA results, the study monitoring committee recommended for an early termination of the study due to lack of efficacy (futility). No further patients were enrolled in the study after the IA.
- The primary analysis of efficacy and all diarrhea-related efficacy endpoints was conducted using the intent-to-treat (ITT) population.
- The superiority of prophylactic OCT versus no prophylactic OCT was assessed using the Pearson Chi-square test in the ITT population.
- The Kaplan-Meier estimates of the proportion of patients experiencing an episode of diarrhea were presented for each 3-week cycle of LAP and CAP by treatment group. Log-rank test was used to compare the treatment groups.

Results

Patient Disposition

- In total, 62 patients were enrolled in this study (OCT+LAP+CAP, n=30; LAP+CAP, n=32) between 17 Dec 2014 (first patient first visit) to 19 Oct 2017 (last patient last visit). Currently, the study is completed, and the present report is the final analysis, which includes all available data (database lock date: 13 Feb 2018).
- In total, 70% of patients in the OCT+LAP+CAP arm and 56% in the LAP+CAP arm completed 8 cycles of the study treatment (24 weeks on study).
- The most common reason for discontinuation of LAP+CAP in both treatment arms was disease progression (OCT+LAP+CAP arm, 73% vs LAP+CAP arm, 63%).
- Two (7%) patients in the OCT+LAP+CAP arm and 3 (9%) in the LAP+CAP arm died due to disease progression.

Demographic and Baseline Characteristics

- The baseline characteristics were well balanced across the 2 treatment arms (**Table 2**).
- The median age was 59 years (range, 33–81) and 55.5 years (range, 36–72 years) in the OCT+LAP+CAP and LAP+CAP treatment arms, respectively.
- All patients except 1 in the LAP+CAP arm had an Eastern Cooperative Oncology Group (ECOG) performance status of 0 or 1 at the time of enrolment in the study.
- Most of the patients presented with visceral and non-visceral metastases in the OCT+LAP+CAP (43%) and LAP+CAP (69%) treatment arms.

Efficacy

- The study did not meet the primary endpoint and was stopped at the IA; prophylactic OCT did not reduce the proportion of patients experiencing grade ≥2 diarrhea. Similar number of patients in both the treatment arms had at least 1 episode of grade ≥2 diarrhea within first 3 cycles (Pearson Chi-square test P value, 0.775) (**Figure 2**).
 - The proportion of patients experiencing at least 1 episode of diarrhea during the entire treatment duration was similar with OCT+LAP+CAP (n=6 [20%]) vs LAP+CAP (n=5 [16%]) (difference: 4.4%; 95% confidence interval [CI]: −20%, 29.2%)
 - A supportive analysis of the primary efficacy using per protocol population also demonstrated similar results as that of the ITT population (difference between the 2 arms: 3.9%; 95% CI: −24.7%, 31.7%)
- Grade ≥3 diarrhea event during the entire treatment period was noted in 2 (7%) patients in the OCT+LAP+CAP arm and none in the LAP+CAP arm.
- Diarrhea events (any grade) during the entire treatment period were higher in the OCT+LAP+CAP arm (n=18; 60%) than in the LAP+CAP arm (n=14; 44%) (difference: 16.3%; 95% CI: −9.3%, 40.4%).
- Median time to onset of the first episode of diarrhea since the start of study treatment in the OCT+LAP+CAP arm was not reached versus 170 days (95% CI: 104; not evaluable [NE]) in the LAP+CAP arm.

Table 2. Demographic and Baseline Characteristics

	OCT+LAP+CAP (n=30)	LAP+CAP (n=32)	Total (N=62)
Median age, years (range)	59 (33–81)	55.5 (36–72)	56.5 (33–81)
Age Categories, n (%)			
≥18 to <65	23 (77)	25 (78)	48 (77)
≥65 to <75	6 (20)	7 (22)	13 (21)
≥75	1 (3)	0	1 (2)
Ethnicity, n (%)			
Hispanic or Latino	0	1 (3)	1 (2)
Non-hispanic or Latino	30 (100)	31 (97)	61 (98)
Race, n (%)			
Asian	0	1 (3)	1 (2)
Caucasian	30 (100)	31 (97)	61 (98)
ECOG performance status, n (%)			
0	18 (60)	13 (41)	31 (50)
1	12 (40)	18 (56)	30 (48)
2	0	1 (3)	1 (2)
Median time since initial diagnosis to randomization, years (range)	3.75 (0.9–13.9)	3.35 (0.7–13.9)	3.50 (0.7–13.9)
Histology at initial diagnosis, n (%)			
Invasive breast carcinoma	21 (70)	15 (47)	36 (58)
Infiltrating carcinoma	5 (17)	9 (28)	14 (23)
Other	1 (3)	7 (22)	8 (13)
Infiltrating lobular carcinoma	1 (3)	0	1 (2)
In situ breast carcinoma	1 (3)	0	1 (2)
Lobular invasive	1 (3)	0	1 (2)
Adenosquamous carcinoma	0	1 (3)	1 (2)
Visceral and/or non-visceral metastases, n	**27**	**30**	**57**
Visceral, n (%)	10 (33)	3 (9)	13 (21)
Non-visceral, n (%)	4 (13)	5 (16)	9 (15)
Visceral and non-visceral, n (%)	13 (43)	22 (69)	35 (56)
Median number of prior anticancer treatment, (range)			
Chemotherapy	4 (3–19)	4 (1–15)	4 (1–19)
Hormonal therapy	1.5 (1–3)	2 (1–5)	2 (1–5)
Biologics	2 (1–10)	1 (1–3)	2 (1–10)
Radiotherapy	2 (1–4)	1 (1–3)	1 (1–4)
ER/PR status, n (%)			
ER+	11 (37)	15 (47)	26 (42)
ER−	19 (63)	17 (53)	36 (58)
PR+	9 (30)	12 (38)	21 (34)
PR−	18 (60)	19 (59)	37 (60)

CAP, capecitabine; ECOG, Eastern Cooperative Oncology Group; ER, Oestrogen or estrogen receptor; LAP, lapatinib; OCT, octreotide; PR, progesterone receptor

Figure 2. Patients Experiencing At least 1 Episode of Diarrhea of Grade ≥2 During the First 3 Cycles of Treatment (ITT)

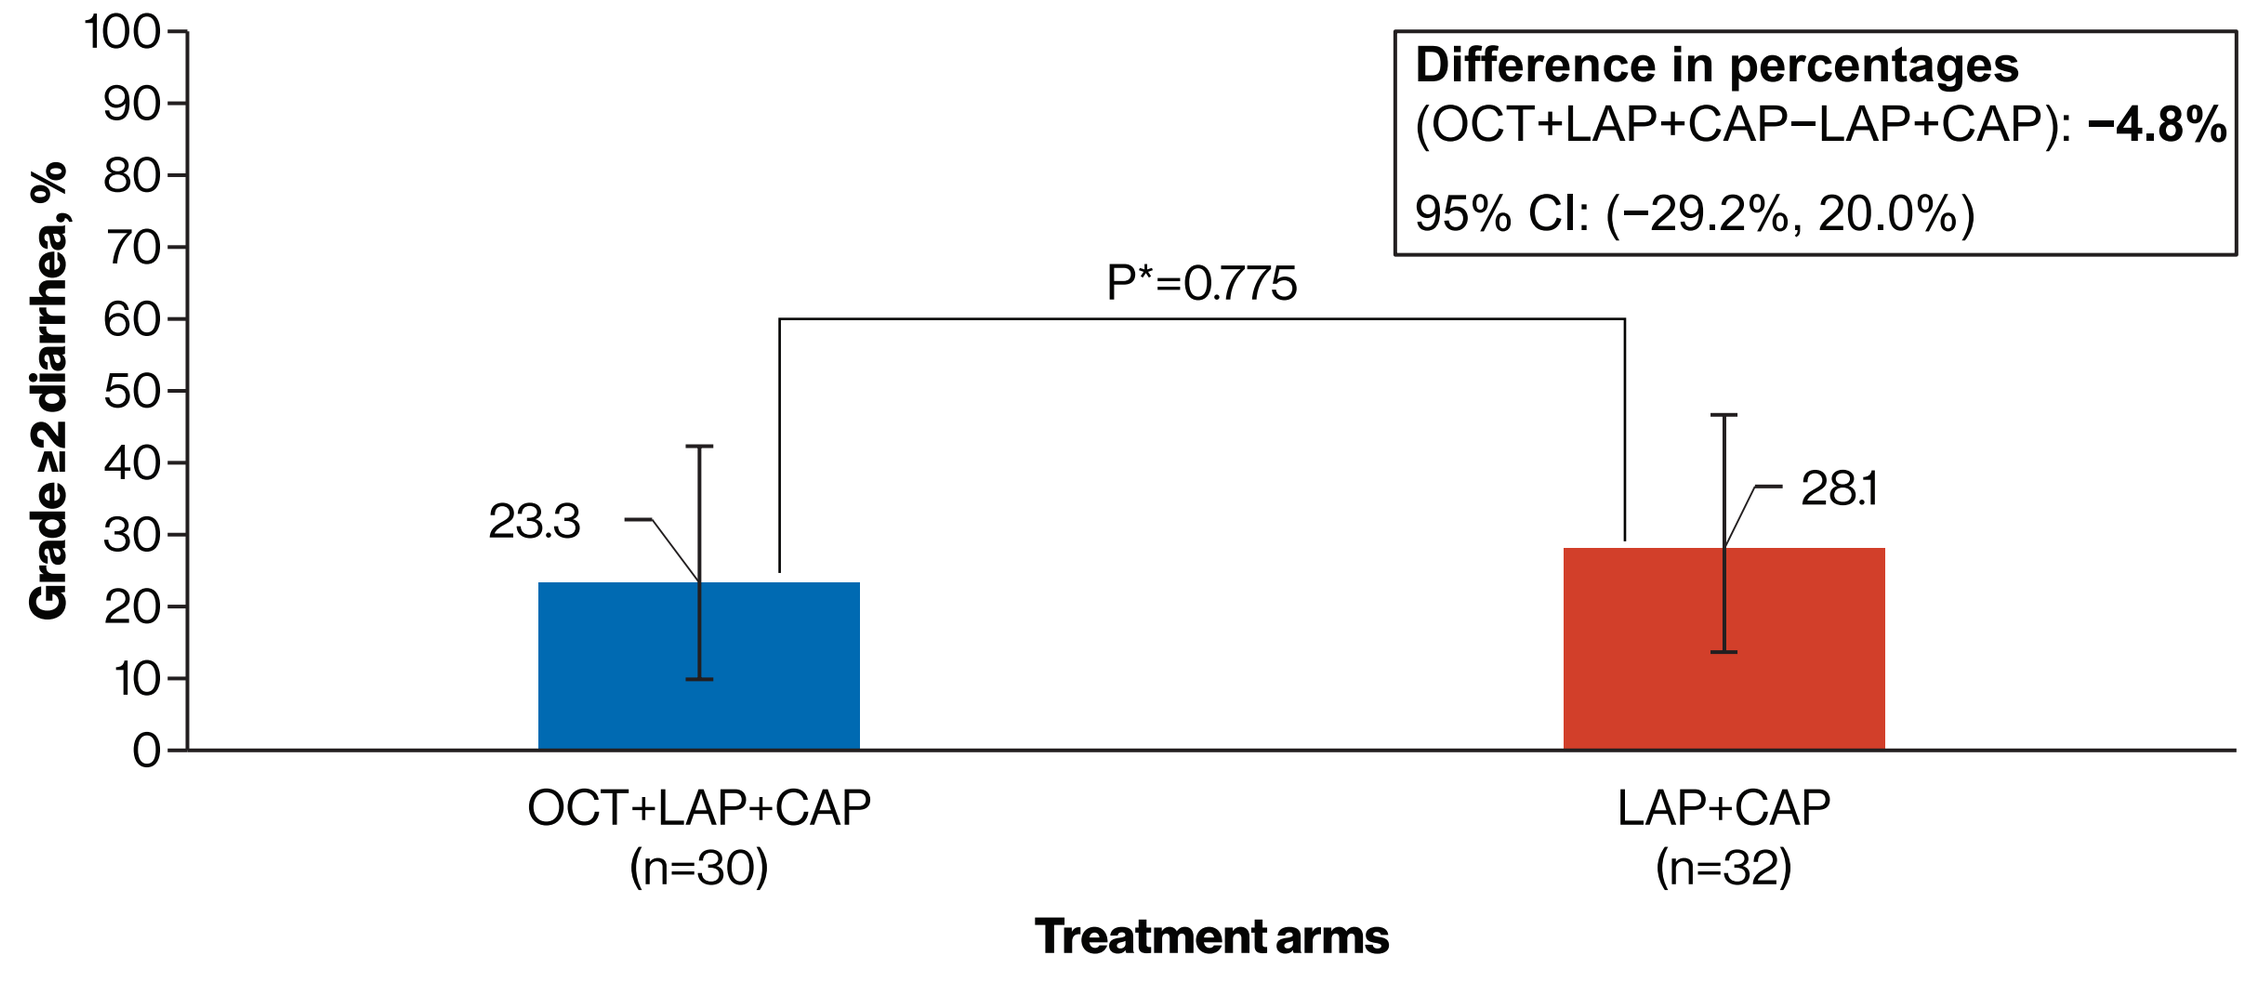

Grade ≥2 diarrhea, %
100
90
80
70
60
50
40
30
20
10
0
Difference in percentages
(OCT+LAP+CAP−LAP+CAP): −4.8%
95% CI: (−29.2%, 20.0%)
P*=0.775
23.3
28.1
OCT+LAP+CAP (n=30)
LAP+CAP (n=32)
Treatment arms

*2-sided P value, Pearson Chi-square test.
CAP, capecitabine; CI, confidence interval; ITT, intent to treat; LAP, lapatinib; OCT, octreotide

- Overall compliance rate was high in both treatment arms (>90%).
- ORR and CBR were similar between the OCT+LAP+CAP and LAP+CAP arms (**Table 3, Figure 3**); 2 (6%) patients achieved CR in the LAP+CAP arm, while none achieved CR in the OCT+LAP+CAP arm.

Table 3. Investigator-assessed ORR and CBR

Best Response, n (%)	OCT+LAP+CAP (n=30)	LAP+CAP (n=32)
ORR (CR+PR)	6 (20)	6 (19)
95% CI	(7.7, 38.6)	(7.2, 36.4)
Difference in ORR% (95% CI)	1.3 (−18.4, 20.9)	
CBR (CR+PR+SD ≥24 weeks)	7 (23)	9 (28)
95% CI	(9.9, 42.3)	(13.7, 46.7)
Difference in CBR% (95% CI)	−4.8 (−26.5, 16.9)	

CAP, capecitabine; CBR, clinical benefit response; CI, confidence interval; CR, complete response; LAP, lapatinib; OCT, octreotide; ORR, overall response rate; PR, partial response; SD, stable disease

Figure 3. Investigator-assessed ORR and CBR

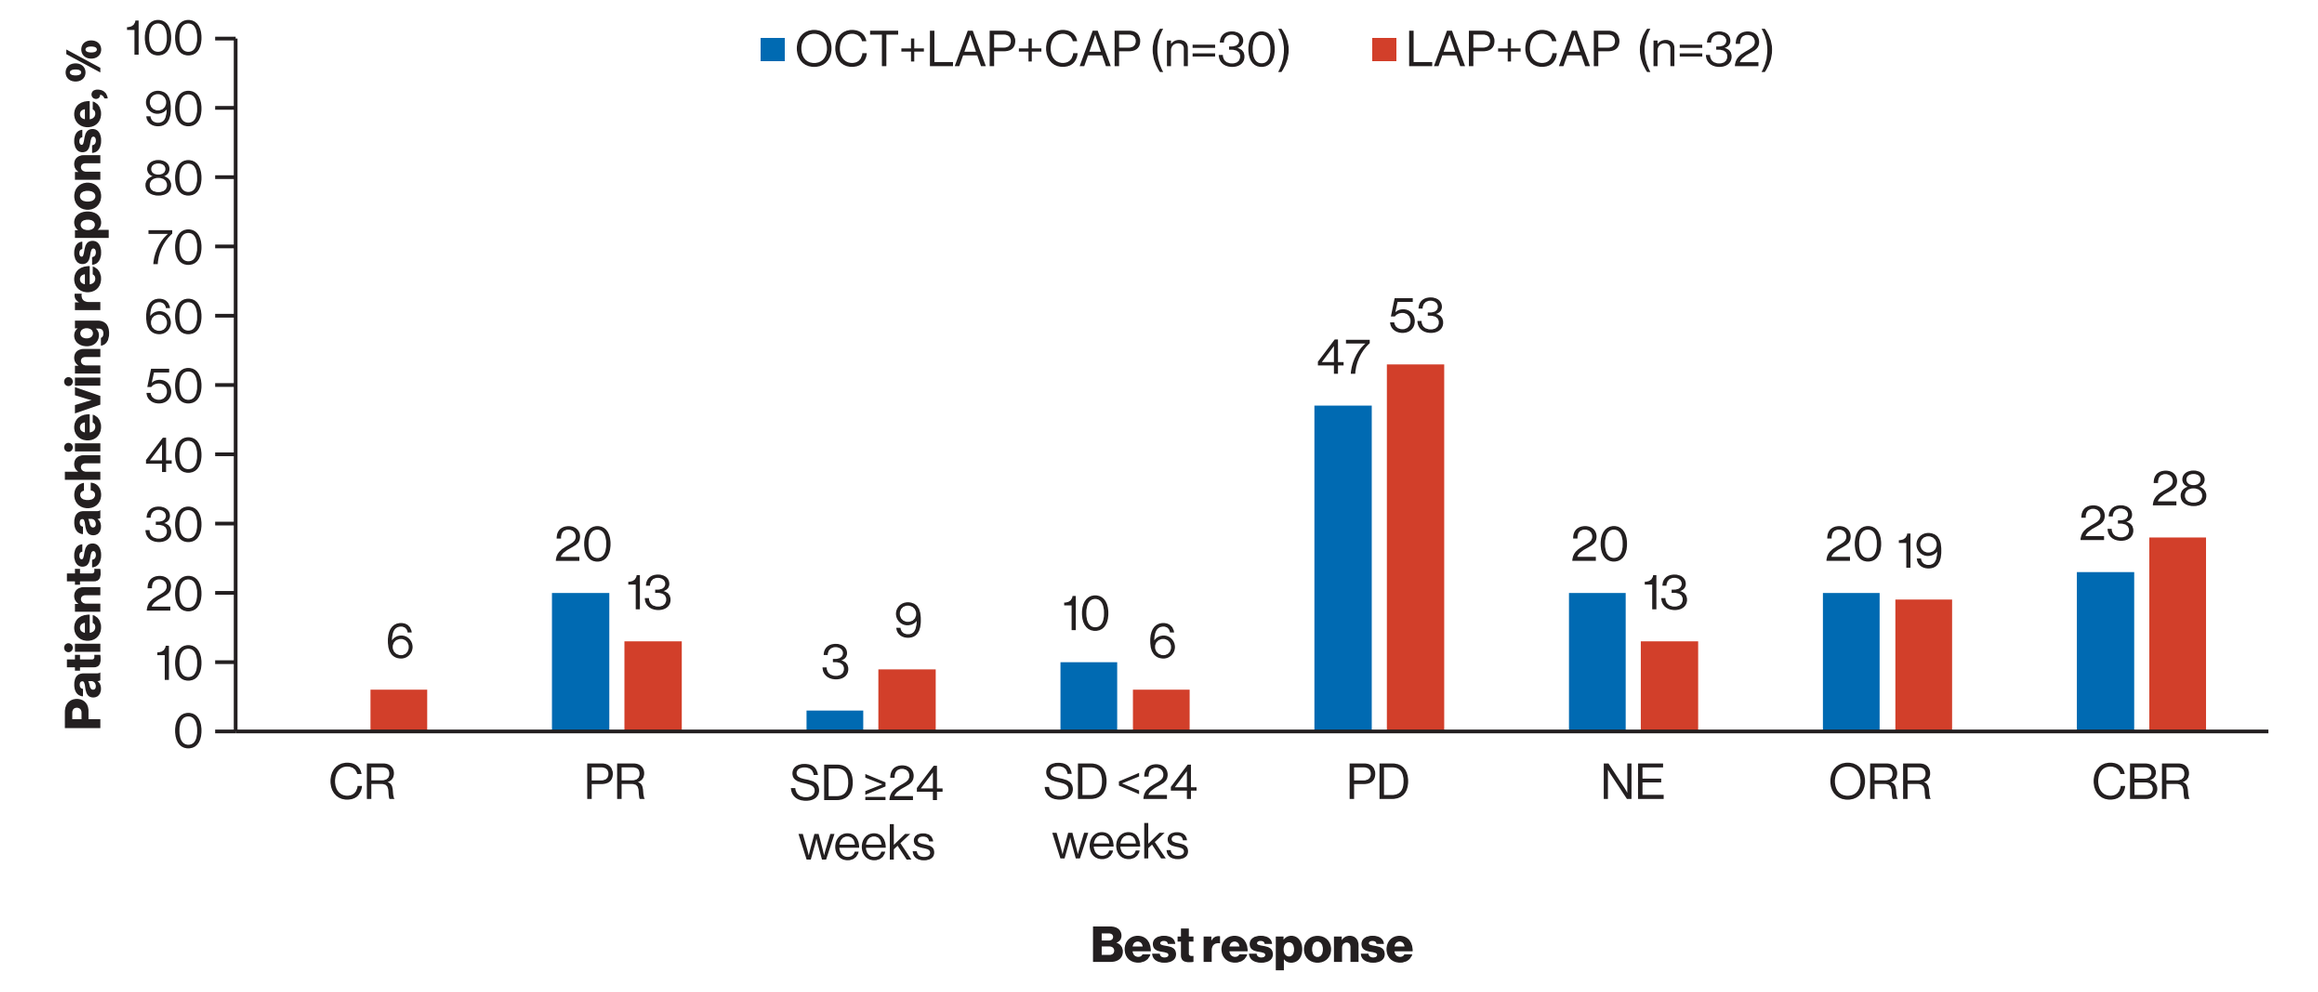

Patients achieving response, %
100
90
80
70
60
50
40
30
20
10
0
OCT+LAP+CAP (n=30)
LAP+CAP (n=32)
6
20
13
3
9
10
6
47
53
20
13
20
19
23
28
CR
PR
SD ≥24 weeks
SD <24 weeks
PD
NE
ORR
CBR
Best response

CAP, capecitabine; CBR, clinical benefit rate; CR, complete response; LAP, lapatinib; NE, not evaluable; OCT, octreotide; ORR, overall response rate; PD, progressive disease; PR, partial response; SD, stable disease

Dosing

- The mean dose intensity of OCT was 75.9 mg.
- LAP dose reductions occurred in 2 (7%) and 3 (9%) patients in the OCT+LAP+CAP and LAP+CAP treatment arms, respectively, mainly due to AEs.
- CAP dose reductions were noted in 12 (41%) patients in the OCT+LAP+CAP arm (mainly due to AEs) and 11 (33%) in the LAP+CAP arm (mainly due to AEs, as per investigator's discretion or non-availability of CAP).
- OCT dose reductions were observed in 2 patients and dose delays in 5 patients.
- Dose interruptions for LAP occurred in 11 patients (37%) in the OCT+LAP+CAP arm and 6 patients (18%) in the LAP+CAP arm, primarily due to AEs. Dose interruptions for CAP occurred in 19 patients (66%) in the OCT+LAP+CAP arm and 16 patients (48%) in the LAP+CAP arm, primarily due to AEs.
- The use of loperamide was more frequent in the LAP+CAP arm (28%) versus the OCT+LAP+CAP arm (13%).

Safety

- Diarrhea was the most frequently observed AE in both treatment arms (59% vs 45%) (**Table 4**).
 - The majority of diarrhea events were of grade 1 or 2 severity and were suspected to be treatment-related events. None of the patients discontinued due to diarrhea
- Hepatobiliary events were noted in 28% versus 24% of patients in the OCT+LAP+CAP versus the LAP+CAP treatment arms.
- The incidence of grade 3/4 AEs was similar between both the treatment arms (31% vs 30%) (**Table 4**).
 - Diarrhea (7%) and palmar-plantar erythrodysaesthesia (7%) were the most frequently observed grade 3/4 AEs in the OCT+LAP+CAP arm, suspected to be treatment-related
- The AEs related to gastrointestinal disorders (66% vs 45%) and skin and subcutaneous tissue disorder AEs (52% vs 45%) were the most frequently observed AEs regardless of study drug relationship in the OCT+LAP+CAP arm versus the LAP+CAP arm, respectively.
- Serious AEs (SAEs) were more frequent in the OCT+LAP+CAP treatment arm (n=5, 17%) versus the LAP+CAP treatment arm (n=3, 9%).
 - Two SAEs (diarrhea and increased alanine aminotransferase [ALT]) in the OCT+LAP+CAP arm and 1 SAE (osteonecrosis of the jaw) in the LAP+CAP arm were suspected to be study drug related
- There were no fatal AEs reported. In total, 5 deaths were reported (OCT+LAP+CAP, 2 [7%]; LAP+CAP, 3 [9%]). There was 1 (3%) on-treatment death in the OCT+LAP+CAP arm due to underlying disease and disease progression.

Table 4. Adverse Events (All grades and Grade 3/4) Regardless of Study Drug Relationship in ≥5% of Patients

Preferred term	OCT+LAP+CAP (n=29)		LAP+CAP (n=33*)	
	All grades n (%)	Grade 3/4 n (%)	All grades n (%)	Grade 3/4 n (%)
Any event	27 (93)	9 (31)	28 (85)	10 (30)
Diarrhea	17 (59)	2 (7)	15 (45)	0
Palmar-plantar erythrodysaesthesia syndrome	13 (45)	2 (7)	11 (33)	2 (6)
ALT increased	4 (14)	1 (3)	2 (6)	0
AST increased	4 (14)	1 (3)	1 (3)	0
Blood bilirubin increased	4 (14)	0	3 (9)	0
Rash	4 (14)	0	7 (21)	0
Fatigue	3 (10)	0	1 (3)	0
Lower respiratory tract infection	3 (10)	1 (3)	1 (3)	0
Thrombocytopenia	3 (10)	0	3 (9)	0
Abdominal pain	2 (7)	0	0	0
Asthenia	2 (7)	0	7 (21)	0
Bilirubin conjugated increased	2 (7)	0	0	0
Blister	2 (7)	0	0	0
Cough	2 (7)	0	3 (9)	0
Dry mouth	2 (7)	0	1 (3)	0
Hyperbilirubinaemia	2 (7)	1 (3)	2 (6)	1 (3)
Nausea	2 (7)	0	2 (6)	1 (3)
Oedema peripheral	2 (7)	0	0	0
Oral candidiasis	2 (7)	0	0	0
Pain in extremity	2 (7)	0	2 (6)	0
Skin fissures	2 (7)	0	2 (6)	0
Stomatitis	2 (7)	0	2 (6)	1 (3)
Vomiting	2 (7)	0	1 (3)	1 (3)
Paronychia	2 (7)	1 (3)	2 (6)	0
Anaemia	1 (3)	0	6 (18)	1 (3)
Decreased appetite	1 (3)	0	2 (6)	0
Lymphoedema	1 (3)	0	2 (6)	1 (3)
Neutropenia	1 (3)	0	4 (12)	0
Bone pain	0	0	2 (6)	1 (3)
Epistaxis	0	0	2 (6)	0
Insomnia	0	0	2 (6)	0
Mouth ulceration	0	0	2 (6)	1 (3)
Oral pain	0	0	2 (6)	0

ALT, alanine aminotransferase; AST, aspartate aminotransferase; CAP, capecitabine; LAP, lapatinib; OCT, octreotide

*N=62 (OCT+LAP+CAP, n=30; LAP+CAP, n=32 is efficacy population) and (OCT+LAP+CAP, n=29, LAP+CAP, n=33 is safety population).

One of the patient received only 0.1 mg OCT dose and did not receive any other OCT dose. Thus, as per safety population definition, this patient was considered under LAP+CAP arm for safety population.

Conclusions

- The addition of prophylactic OCT did not show a statistically significant or clinically meaningful reduction in the occurrence of grade ≥2 diarrhea in the patient population. The study was stopped at the IA due to futility.
- Diarrhea (overall, all grades), which is different from primary endpoint (grade ≥2 diarrhea), was more frequent in patients in the OCT+LAP+CAP arm compared with those in the LAP+CAP arm.
- Loperamide was more frequently used in patients not receiving OCT compared with those receiving OCT.

References

1. Ryan Q, et al. *Oncologist*. 2008;13(10):1114-1119.
2. Tykerb® Prescribing Information. 2017.
3. Gamucci T, et al. *J Cancer Res Clin Oncol*. 2014;140:221-226.
4. Benson A, et al. *J Clin Oncol*. 2004;22:2918-2926.
5. UpToDate® website. https://www.uptodate.com/contents/management-of-acute-chemotherapy-related-diarrhea. Accessed 4 October 2018.
6. Octreotide LAR Summary of Product Characteristics (eMC) January 2014.

Acknowledgements

Original funding for the RELATED study was provided by GlaxoSmithKline; as of 2 March 2015, LAP is an asset of Novartis Pharmaceuticals Corporation, the current sponsor of this study. Novartis Pharmaceuticals Corporation provided financial support for the medical editorial assistance. We thank Bhavani Yamsani, MPharm, MBA (Novartis Healthcare Private Ltd) for the medical editorial assistance with this poster.

Poster Presented at the European Society for Medical Oncology (ESMO), 19–23 October 2018, held at Munich, Germany.

Text: Qf37b4
To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe & Russia
+46737494608 Sweden, Europe
Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=f37b4

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Scan this QR code

corresponding author
(dr.krivorotko@mail.ru)